The use of a novel 'active' inhalation device to deliver high respirable fractions of high dose dry powder active agents to the lung.

Paul M Young*[1], Jim Thompson[1,2],Robert Price[1], Derek Woodcock[2], Keith Davies[2]

[1]Pharmaceutical Technology Research Group, Department of Pharmacy and Pharmacology, University of Bath, UK.
[2]Britannia Pharmaceuticals, Redhill, Surrey, UK.

INTRODUCTION

The use of dry powder inhalers (DPI) for the delivery of medicaments for respiratory disease has become popular in recent years. Such a move is mainly due to both the ease of use and chemical stability of the active in a solid form. Conventional DPI devices are based around the delivery of relatively low doses (<400 μg) of micron-sized material (<5 μm diameter). However to date, the delivery of higher doses applicable, for example, to vaccine delivery have not been investigated.

As part of an ongoing development program, a novel 'active' dry-powder inhalation PADD device (pressurised aerosol dry-powder delivery) has been developed for delivery of high dose (10-1000 mg) cohesive powders to the respiratory tract.

Preliminary in vitro studies have been undertaken using Pumactant™, an artificial lung surfactant, which has been shown to have a prophylactic effect (100 mg) in recent clinical trial studies[1]. Pumactant™ is a mixture, of dipalmatidylphosphatidylcholine and phosphatidylglycerol (DPPC and PG) and has similar properties to that of indigenous surfactant. However, due to such similarity, it has a low transition temperature, high affinity for moisture and is therefore is naturally cohesive. Clearly an ideal subject for study using the PADD active delivery system.

MATERIALS AND METHODS

Micronised Pumactant™ was first characterised for particle size and morphology. Particle size distribution was obtained by laser light scattering (Malvern Mastersizer X, Malvern UK) using a small volume dispersion cell and cyclohexane as a dispersant (samples were sonicated for 5 mins prior to analysis). Particle morphology was investigated using scanning electron microscopy at 10kV (JEOL 6310, Jeol, Japan).

A schematic diagram of the PADD device is shown in Figure 1. The PADD device is a handheld high-energy aerosolisation system utilising pressurised CO_2 gas (in this case) to supply a positive pressure (through a series of Venturi tubes) into a carrier-free powder bed. The aerosolised powder is entrained and inhaled through a conventional mouthpiece. Can pressure for the initial studies was 12bar, which lasted a duration of approximately 7 seconds.

For comparative purposes, aerosolisation efficiency of the active PADD device was compared to an off the shelf conventional DPI (Cyclohaler™), using a twin stage impinger[2] (Copley Scientific, Nottingham, UK) at 60L.min^{-1}. Delivered dose and repeatability were investigated using the dose unit sampling apparatus[2] (DUSA) (Copley) at 30L.min^{-1}, while the influence of delivered dose on aerosol size distribution was investigated using the Anderson cascade impactor[2] (ACI) (Copley).

Drug content, obtained from the deposition studies were calculated by mass and HPLC
(using a evaporating light scattering detector and validated method).

A representative photograph of the PADD active dry powder Inhaler device, with conventional mouthpiece, CO_2 canister and large fill sample vial are shown in Figure 2 alongside a dissembled ACI with dose deposition post testing (with a ~100 mg Pumactant™ dose).

Figure 2 (right). Photographic image of the PADD aerosolisation device assembly and particle deposition pattern on Anderson cascade plates post analysis (~ 100 mg dose)

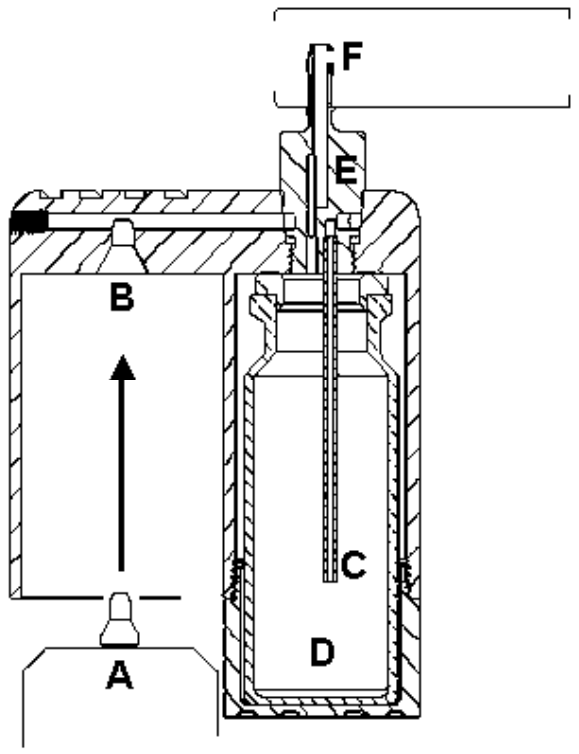
F
E
B
C
D
A

Figure 1. Schematic diagram of the PADD dry powder aerosolisation device. Where A. pressurised CO_2 canister, B. connection duct and transfer conduit, C. aerosolisation pipe, D. vial containing drug material, E. exit channel, F. PADD orifice and patient mouthpiece.

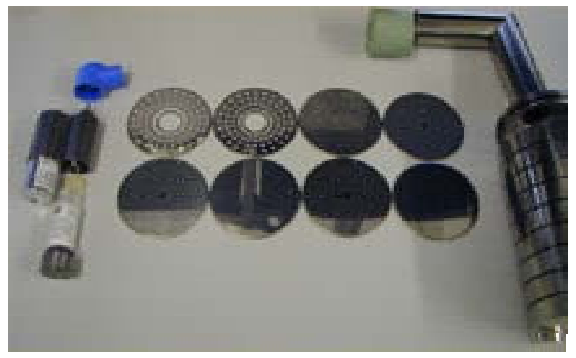

RESULTS AND DISCUSSION

A representative SEM image of the Pumactant™ with particle size distribution is shown in Figure 3. The micronisation process was conducted at low temperatures to avoid thermal transitions and the subsequent particulates appeared to be discrete entities with a median diameter of 1.49 μm ± 0.06 μm (n=3). However, the SEM images suggested the material to be highly agglomerated. Aerosolisation efficiency, measured using the a conventional Cyclohaler™ indicated that although the total delivery efficiency was relatively high (>60% for a 50mg loaded dose) no fine particle fraction of Pumactant™ was recovered from stage 2 of the TSI. Such observations suggest that the energy imparted by a non-active device is not strong enough to overcome particle cohesion. In comparison, initial TSI investigations using an non-optimised PADD device indicated a delivery efficiency of 64 % ± 21% with a FPF of 39% ± 14%.

Optimisation of the PADD device led to improved delivery efficiency and increased fine particle fractions. Depending on target dose, delivery efficiency was between 60 and 70% with relative standard deviations less than 6% (n>5).

Particle deposition profiles (Figure 4) obtained by ACI suggested similar particle deposition profiles (15, 33, 50 and 75 mg delivered dose), with a mean fine particle fraction (particles < 3.3 μm) across the delivered dose range of 54% ± 10%.

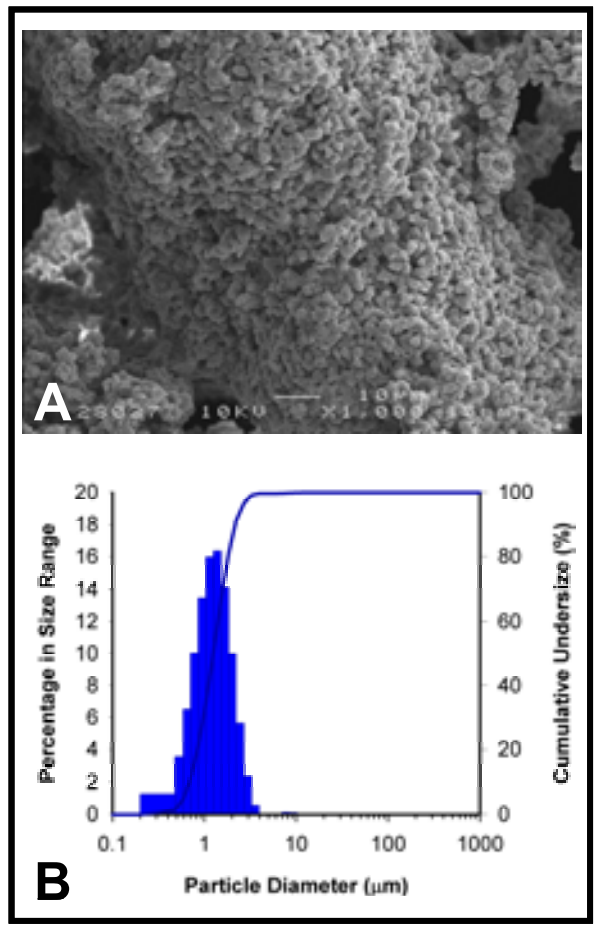
A
B
Percentage in Size Range
Cumulative Undersize (%)
Particle Diameter (μm)
0.1
1
10
100
1000

Figure 3. (A) Scanning electron micrograph of the micronised Pumactant.™ (B) Particle size distribution,

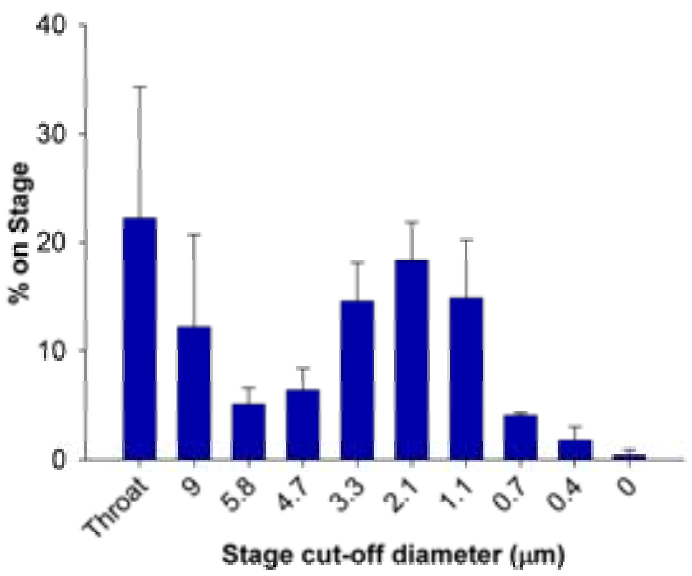
% on Stage
Stage cut-off diameter (μm)
Throat
9
5.8
4.7
3.3
2.1
1.1
0.7
0.4
0

Figure 4. Mean percentage recovery for ACI deposition

Conclusions

The delivery of high dose medicaments to the respiratory tract is a technology yet to be realised. Initial studies have indicated that when considering non-active devices the energy required to efficiently aerosolise such doses may not be feasible.

Preliminary studies, using a high dose asthma therapy drug, (Britannia's Pumactant™) and active dry powder inhaler (Britannia PADD device) have shown such goals to be possible.

References

1. Babu,K.S., Woodcock, D.A., Smith, S.E., Heminsley, A.M., Little, L., Staniforth, J.N., Holgate, S.T., and Conway, J.H. Pumactant abolishes early asthmatic response in patients with allergic asthma, Proceedings of the American Thoracic Society (2002)

2. Description of the TSI, DUSA and ACI apparatus can be found in the British Pharmacopoeia. Methods used through out the study followed the specific guidelines.

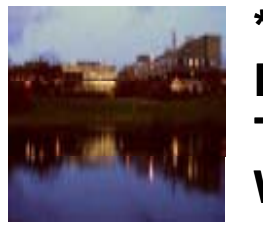

***Correspondence: Paul M Young**
Email: prspmy@bath.ac.uk
Tel:+44 (0) 1225 383103
Web:http://www.bath.ac.uk/~prspmy/